Physician Concordance With Update to ASCO Guidelines for Antiemetic Use With Carboplatin AUC ≥ 4

ASCO 2019,
May 31 – June 4, 2019

Rudolph M. Navari[1], Kathryn J. Ruddy[2], Thomas W. LeBlanc[3], Rebecca Clark-Snow[4], Rita J. Wickham[5], Gary Binder[6], Tammy Coberly[6], Ravi Potluri[7], Luke M. Schmerold[7], Eric J. Roeland[8]

[1]University of Alabama Birmingham School of Medicine; [2]Division of Medical Oncology, Mayo Clinic; [3]Duke Cancer Institute; [4]Oncology Consultant; [5]Rush University College of Nursing; [6]Helsinn Therapeutics US, Inc.; [7]SmartAnalyst, Inc.; [8]Massachusetts General Hospital

BACKGROUND

In 2017, the National Comprehensive Cancer Network (NCCN) (2/2017) and ASCO (8/2017) each amended antiemetic guidelines to classify carboplatin AUC ≥ 4 as a highly emetogenic chemotherapy (HEC), recommending upfront triple prophylaxis with an NK1 receptor antagonist (RA) + 5-HT3 RA + dexamethasone.[1,2] This change aligned these guidelines with previous recommendations made in March 2016 by the Multinational Association for Supportive Care in Cancer (MASCC). Previously, carboplatin was considered moderately emetogenic.

Effective January 2018, the US Centers for Medicare and Medicaid Services (CMS) instituted an oncology quality outcome measure, OP-35. This measure assesses "potentially avoidable" acute care, defined as 30-day post-chemotherapy emergency department (ED) or inpatient admission (IP) use associated with nausea and vomiting (NV) or any of eight other common chemotherapy-related toxicities.

OBJECTIVES

- To identify and characterize antiemetic prophylaxis for courses of carboplatin and compare with courses of other HECs, oxaliplatin, other non-HECs, and HEC/MEC orals.
- To assess the impact of the recent addition of carboplatin as HEC within antiemetic prophylaxis guidelines.
- To assess the acute care utilization by patients following receipt of carboplatin, relative to other chemotherapies with similar emetogenic potential.

METHODS

- DATA SOURCE
 - We analyzed data from the IBM Watson Health™ Explorys electronic health record (EHR) database consisting of 40 large US Integrated Delivery Networks with approximately 55 million patients.
 - We selected patients initiating chemotherapy from October 2012 through August 2018 using drug procedure codes and categorized groups by emetogenic potential as defined by NCCN.
 - We specifically evaluated carboplatin, other highly emetogenic chemotherapy (HECs), oxaliplatin, non-HEC IV chemotherapy, and highly/moderately emetogenic (HEC/MEC) oral chemotherapy. Oxaliplatin was included to determine any differences in outcomes relative to carboplatin, given its platinum content yet categorization as non-HEC.
 - For each patient receiving chemotherapy, we defined a distinct course of chemotherapy as an initial administration followed by periodic subsequent administrations (multiple cycles) that continued until a 90-day treatment gap occurred (maximum of 180 days).
 - A 90-day chemotherapy-free period was required prior to the initial administration.
 - We included only carboplatin courses with ≥14 day-cycles (as a proxy for AUC ≥ 4) and other HEC courses with ≥7 day-cycles.
 - At least 30 days of data availability after the start of the course of therapy was required.
 - We included only adult patients (≥18 years old at the start of the course of therapy).
- OUTCOMES ASSESSED
 - Rate of antiemetic prophylaxis guideline adherence for carboplatin and other categories of chemotherapy, defined as administration of triple prophylaxis (NK1 RA + 5HT3 RA + dexamethasone) at chemotherapy initiation.
 - Rate of NV observed in any care setting during the course of therapy (ICD-9-CM 307.54, 535.41, 536.2, 578.0, 787.01-787.04; ICD-10-CM F50.8, K29.61, K92.0, R11.0, R11.1x, R11.2).
 - Rate of "potentially avoidable" acute care as defined by OP-35, the oncology outcome measure recently implemented by CMS[4], specifically:
 - 30-day post-chemotherapy acute care (IP or ED use), involving nausea or vomiting (NV) or any of eight other toxicities deemed potentially avoidable (anemia, dehydration, diarrhea, fever, neutropenia, pain, pneumonia, or sepsis). The acute care could occur within 30 days after the start of any cycle during the course of that chemotherapy.
 - This measure is specific to chemotherapy administered in the outpatient setting to non-leukemia patients; the 30-day acute care and OP-35 analyses were done on this subset of courses (83% of overall courses and 89% of carboplatin courses).
 - All data management and analyses were done using SAS 9.4 and Microsoft Excel 2016 programs.

RESULTS

- We identified 92,052 courses of the chemotherapy groups of interest that met the respective inclusion criteria **(Fig. 1)**.
- A descriptive analysis of demographic characteristics is shown in **Table 1**. 70% of carboplatin courses were given to females; females received 43-61% of the other chemotherapy groups.
- Before the guideline change, rates of upfront triple prophylaxis with carboplatin grew from 14% in 2013 to 16% in mid-2017.
- Rates then rose to 26% by 1Q 2018 before dropping to 20% by 3Q 2018; quarterly rates averaged 20% (range 15%-26%) following the guideline change **(Fig. 2)**.
- In 31% of carboplatin courses we noted 30-day acute care use, of which 75% involved ≥1 of the ten OP-35 toxicities **(Fig. 3)**.
- NV (with or without acute care use) was reported in 24% of courses, and 27% of total OP-35 acute care events involved NV **(Figs. 3,4)**.
- Rates for NV, and for OP-35-related and NV-related acute care after carboplatin, were similar to rates after other HEC chemotherapy, and higher than rates after other non-HEC IV chemotherapy or oral HEC/MEC agents. Rates for oxaliplatin were also similar to HEC **(Fig 4)**.

Figure 1. Attrition Diagram – Courses of Chemotherapy

Total instances of chemotherapy administration from October 2012 – August 2018

N = 1,425,984

↓

Distinct chemotherapy courses with no chemotherapy use during 90 days preceding initiation

Carboplatin	Other HECs	Oxaliplatin	Other non-HECs	HEC/MEC Oral
n = 16,938	n = 20,244	n = 5,729	n = 60,191	n = 2,111

↓

Courses with ≥14-day cycles for carboplatin and ≥7-day cycles for oxaliplatin and other HECs

Carboplatin	Other HECs	Oxaliplatin	Other non-HECs	HEC/MEC Oral
n = 11,956	n = 16,533	n = 4,975	n = 60,191	n = 2,111

↓

Courses with ≥30 days of data available following initiation of chemotherapy

Carboplatin	Other HECs	Oxaliplatin	Other non-HECs	HEC/MEC Oral
n = 11,587	n = 16,253	n = 4,877	n = 58,528	n = 2,068

↓

Courses for patients at least 18 years old at initiation of chemotherapy

Carboplatin	Other HECs	Oxaliplatin	Other non-HECs	HEC/MEC Oral
n = 11,554	n = 16,025	n = 4,876	n = 57,556	n = 2,041

HEC: highly emetogenic chemotherapy; MEC: moderately emetogenic chemotherapy

Figure 2. Carboplatin Triple Antiemetic Prophylaxis Use Over Time

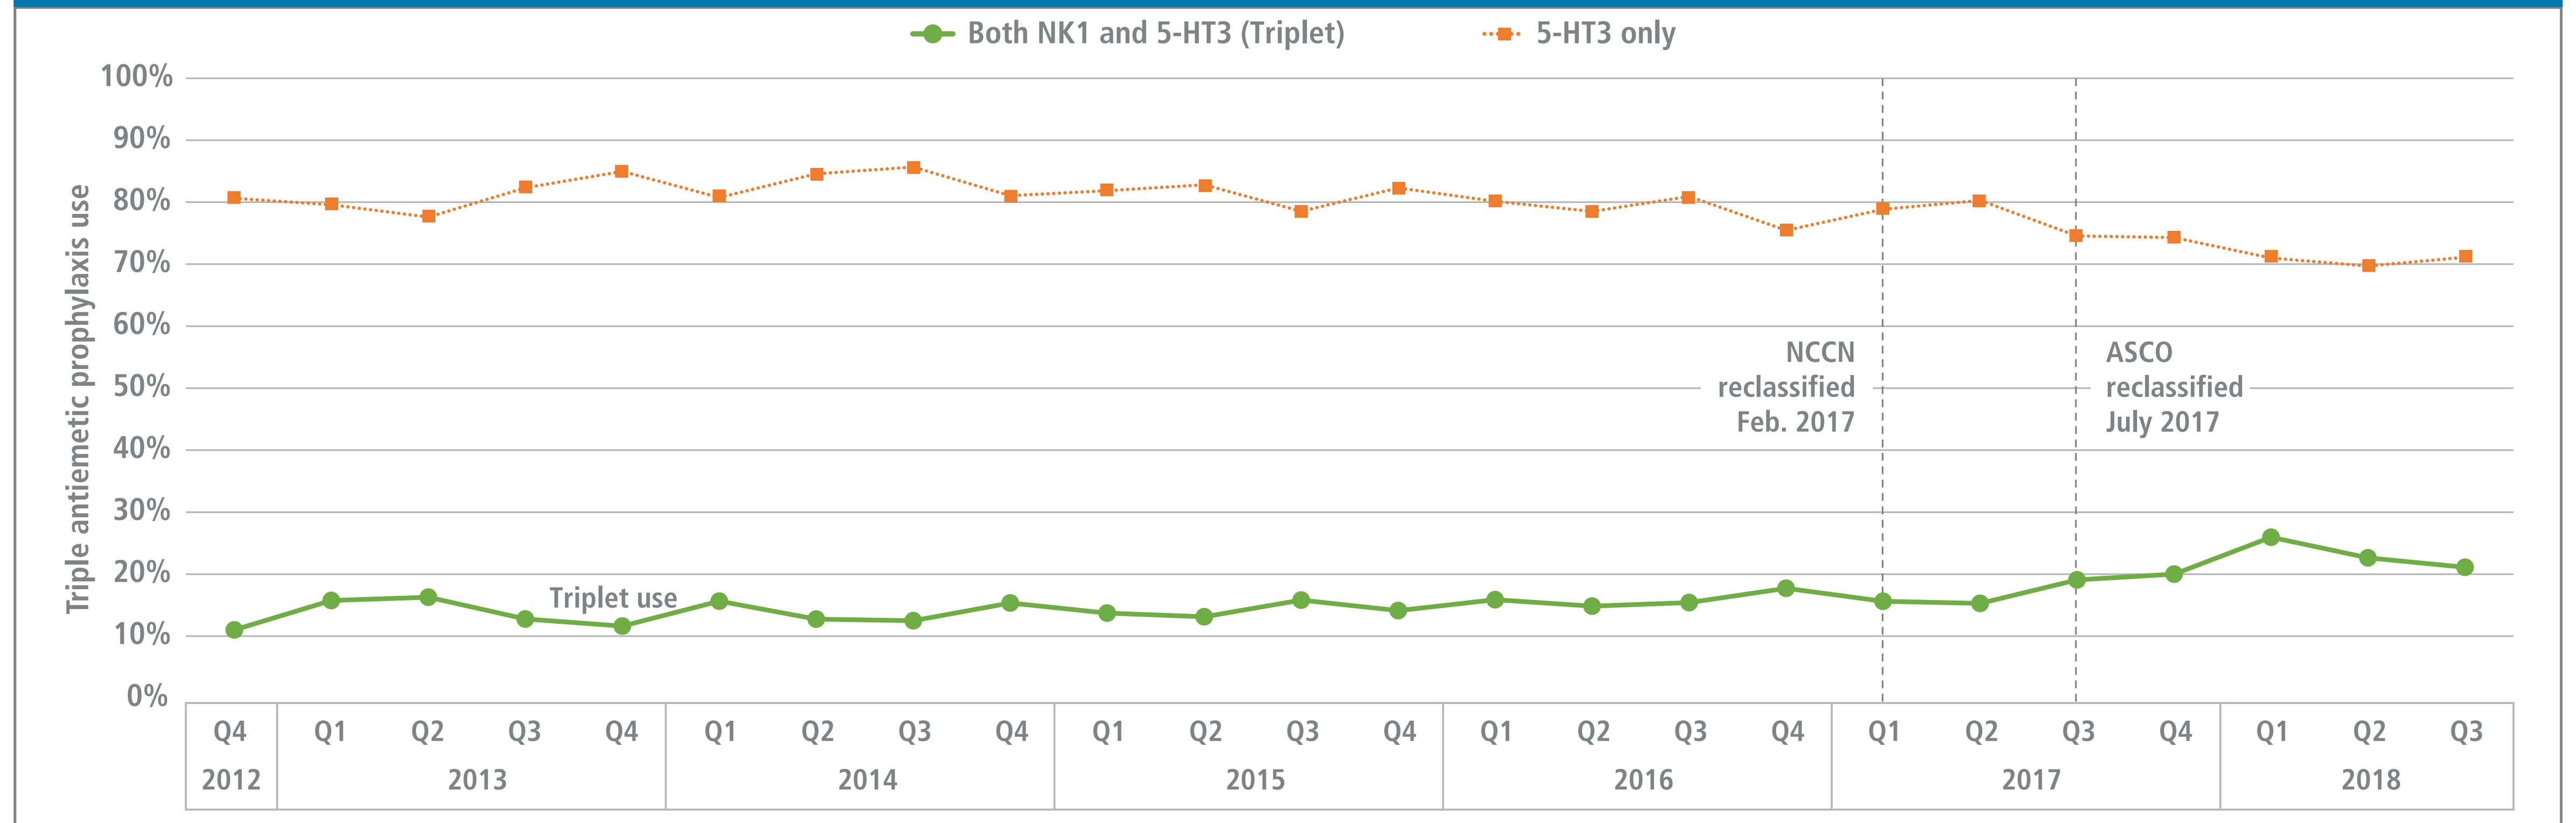
Both NK1 and 5-HT3 (Triplet)
5-HT3 only
Triple antiemetic prophylaxis use
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Triplet use
NCCN reclassified Feb. 2017
ASCO reclassified July 2017
Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3
2012
2013
2014
2015
2016
2017
2018

Note: Dexamethasone use occurred with virtually all patients in addition to the above-described prophylaxis.

Table 1. Demographic Characteristics

	Carboplatin	Other HECs	Oxaliplatin	Other non-HECs	HEC/MEC Oral
Age, years					
Mean (SD)	64 (12)	60 (13)	62 (12)	63 (15)	60 (14)
Median (Q1–Q3)	66 {57 - 73}	61 {52 - 69}	62 {54 - 70}	65 {55 - 74}	62 {52 - 70}
Female, n (%)	8,125 (70%)	9,827 (61%)	2,096 (43%)	33,353 (58%)	1,099 (54%)
Region, n (%)					
Midwest	7,773 (67%)	9,933 (62%)	3,089 (63%)	34,502 (60%)	1,431 (70%)
South	2,087 (18%)	3,362 (21%)	973 (20%)	12,066 (21%)	283 (14%)
West	1,467 (13%)	2,397 (15%)	727 (15%)	9,435 (16%)	275 (13%)
Northeast	161 (1%)	236 (1%)	54 (1%)	1,205 (2%)	44 (2%)
Unknown	66 (1%)	97 (1%)	33 (1%)	348 (1%)	8 (0%)
Race, n (%)					
White	9,277 (80%)	12,379 (77%)	3,824 (78%)	45,799 (80%)	1,760 (86%)
Black	1,208 (10%)	1,777 (11%)	528 (11%)	5,750 (10%)	127 (6%)
Multi-racial	455 (4%)	795 (5%)	218 (4%)	2,683 (5%)	102 (5%)
Other	223 (2%)	474 (3%)	117 (2%)	1,124 (2%)	26 (1%)
Unknown	391 (3%)	600 (4%)	189 (4%)	2,200 (4%)	26 (1%)
Cancer type, n (%)					
Breast	2,032 (18%)	5,217 (33%)	93 (2%)	5,105 (9%)	131 (6%)
Lung	4,185 (36%)	1,615 (10%)	112 (2%)	3,703 (6%)	173 (8%)
GI	1,095 (9%)	2,009 (13%)	4,601 (94%)	7,045 (12%)	74 (4%)

GI: Gastrointestinal; HEC: highly emetogenic chemotherapy; IV: intravenous; MEC: moderately emetogenic chemotherapy

Figure 3. Potentially Avoidable Acute Care Events After Carboplatin

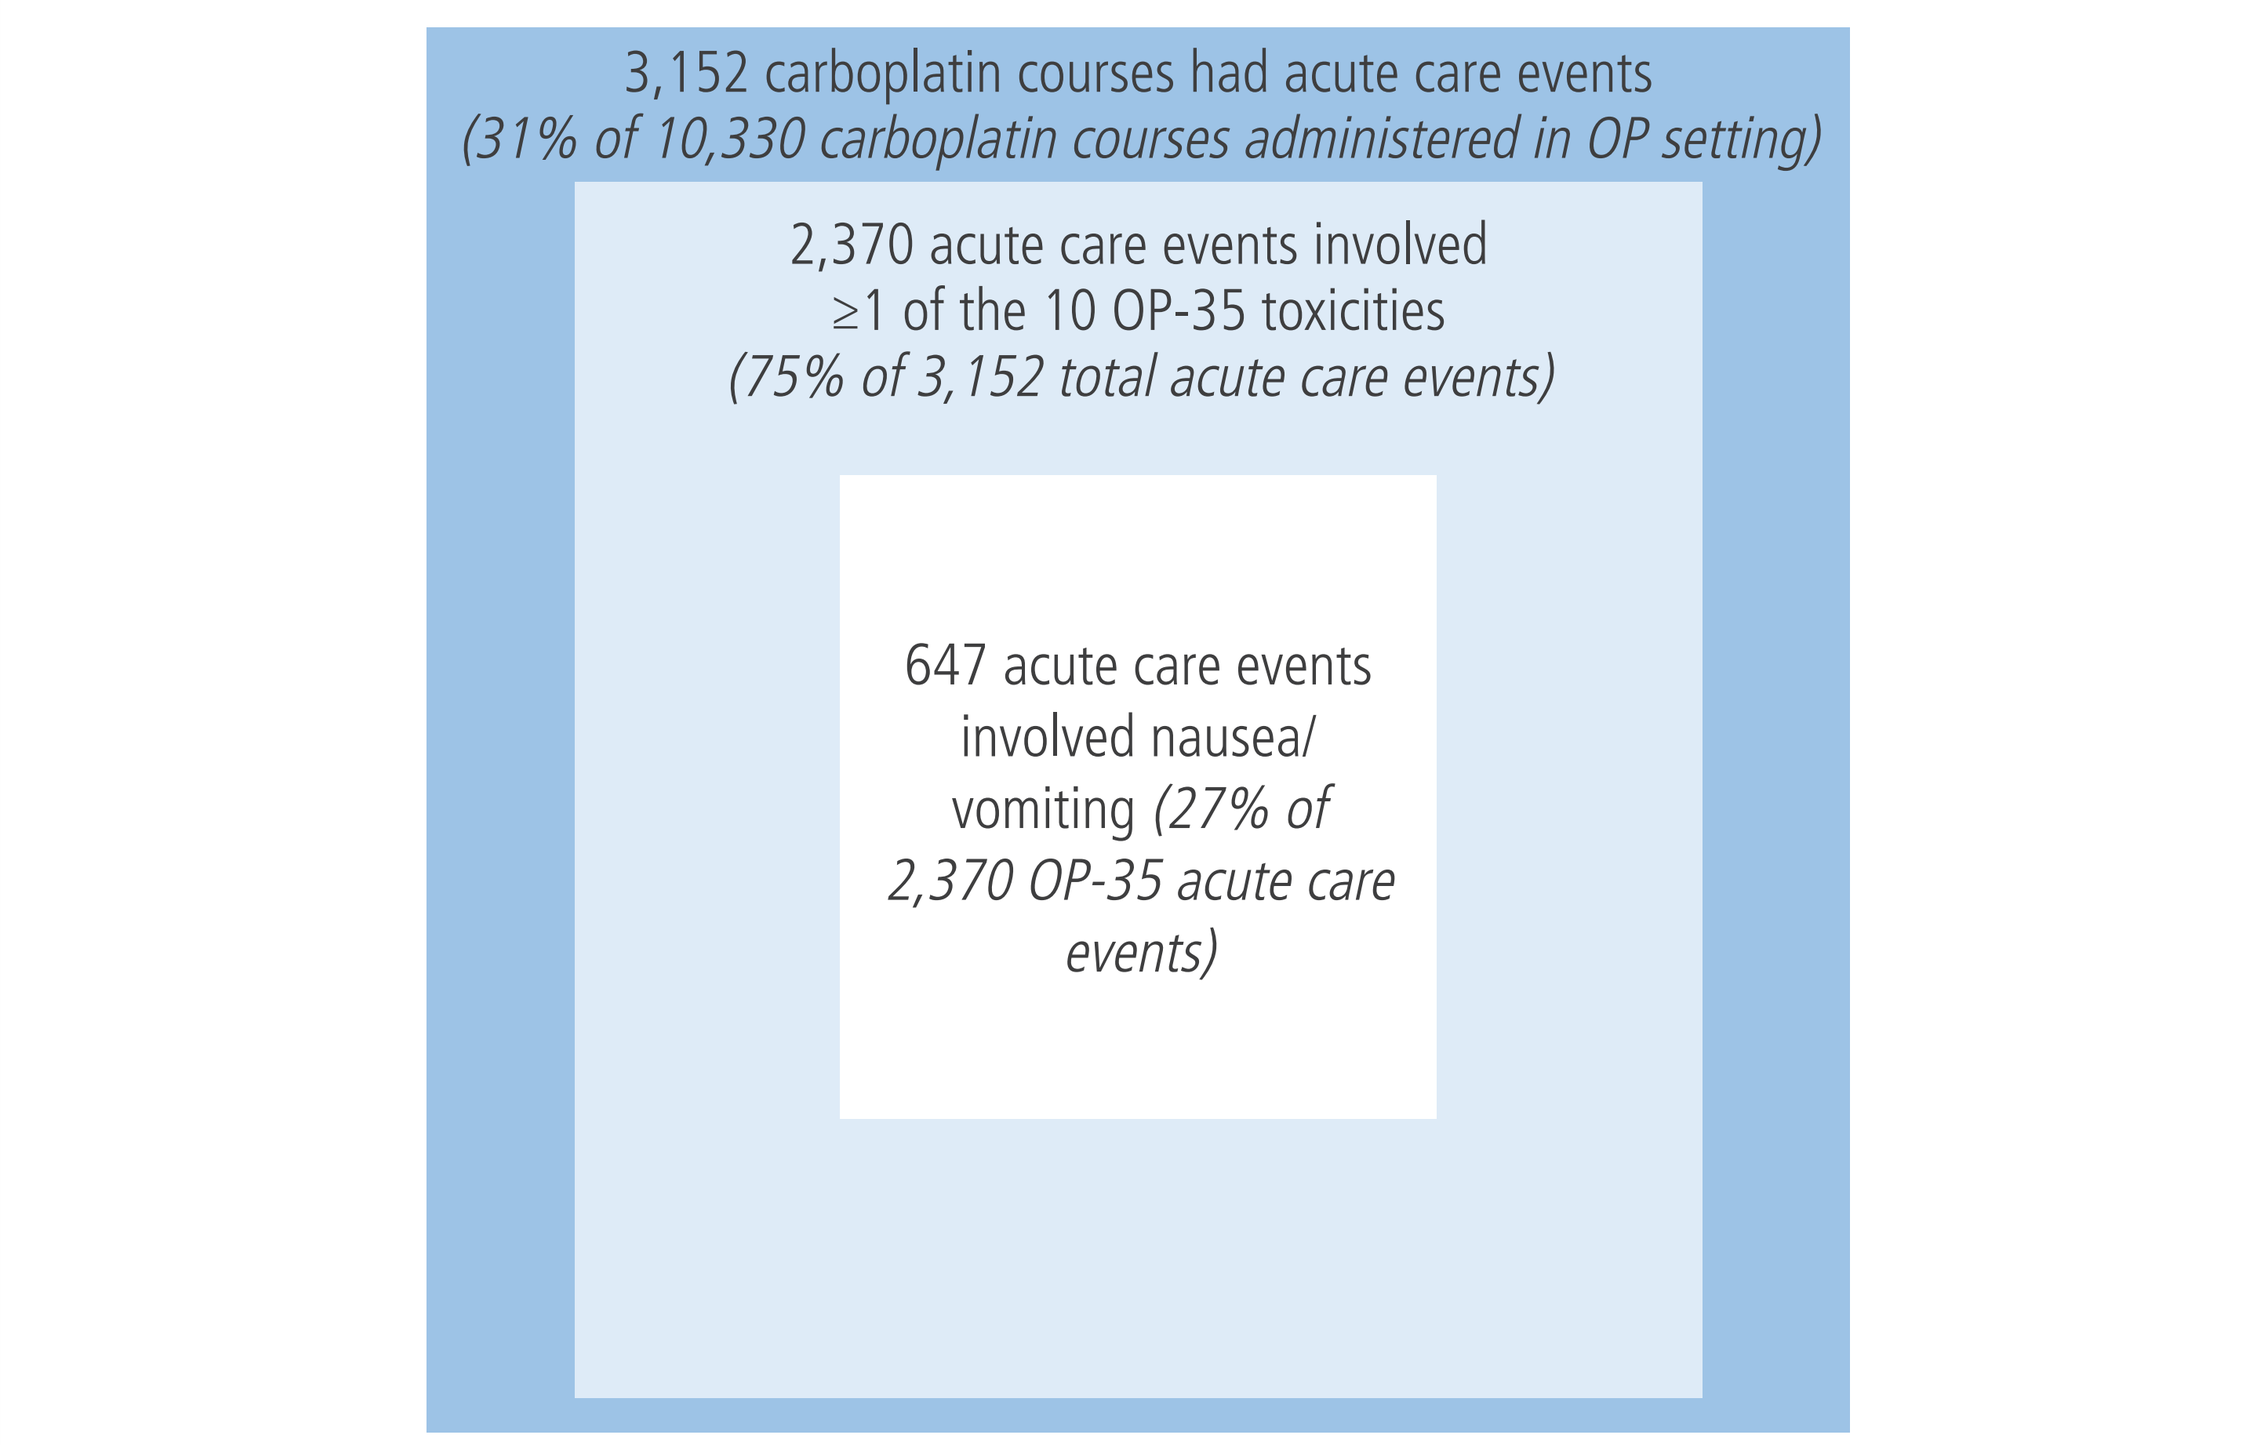
3,152 carboplatin courses had acute care events
(31% of 10,330 carboplatin courses administered in OP setting)
2,370 acute care events involved
≥1 of the 10 OP-35 toxicities
(75% of 3,152 total acute care events)
647 acute care events involved nausea/vomiting (27% of 2,370 OP-35 acute care events)

Note: The area of each box is proportional to the percentage listed.

DISCUSSION

- This data suggests incomplete awareness of the addition of carboplatin to the HEC category and/or physician belief that triple antiemesis prophylaxis is not required for carboplatin. In addition, past research indicates considerable gaps in antiemetic prophylaxis even for cisplatin,[5,6] which is universally recognized as HEC. We hypothesize that reasons for these gaps in use of antiemetic prophylaxis include the following: subordination of physician attention to supportive care relative to therapeutic intervention, physicians neither seeking nor receiving reports of actual CINV symptoms from patients, and clinical inertia.[7] In some cases, these may be exacerbated by cost pressure to avoid use of NK1 agents.
- Limitations of this analysis include the use of EHR data, which may understate certain symptoms and understate acute care provided at non-network sites.
- We suggest the following to resolve this shortcoming in care:
 - Ensure oncologist involvement in institutional or practice formulary committees deciding on chemotherapy "order sets" which determine the associated antiemetic prophylaxis and other supportive care medications within electronic order systems.
 - Assign a "champion" within each practice or institution to lead regular reviews of antiemetic guidelines, ensure order sets follow updated antiemetic guideline recommendations, and actively measure and manage physician adherence. (There is no evidence supporting a "watch and wait" approach to prophylaxis).
 - Continue to increase awareness regarding the increasing role of oncology outcome measures (such as CMS' OP-35) and association to future reimbursement.
 - Consider the use of the most advanced and recently studied antiemetic prophylaxis agents and evaluate the economics on "total cost of care" and quality of outcomes, without regard for impact on the pharmacy budget relative to medical budget.
 - Develop strategies to monitor patients' common chemotherapy-induced symptoms between chemotherapy visits and take action as warranted; this has been shown to improve overall survival.[8,9]
- The need for substantially greater physician concordance with antiemetic guidelines for carboplatin is plainly signaled by the high level of avoidable acute care use involving CINV. Improvement in rates of triple prophylaxis may be one of the most feasible ways to reduce a costly and meaningful toxicity and preclude poor quality ratings and negative reimbursement impacts from CMS.

Figure 4. Nausea/Vomiting and Potentially Avoidable Acute Care by Chemotherapy

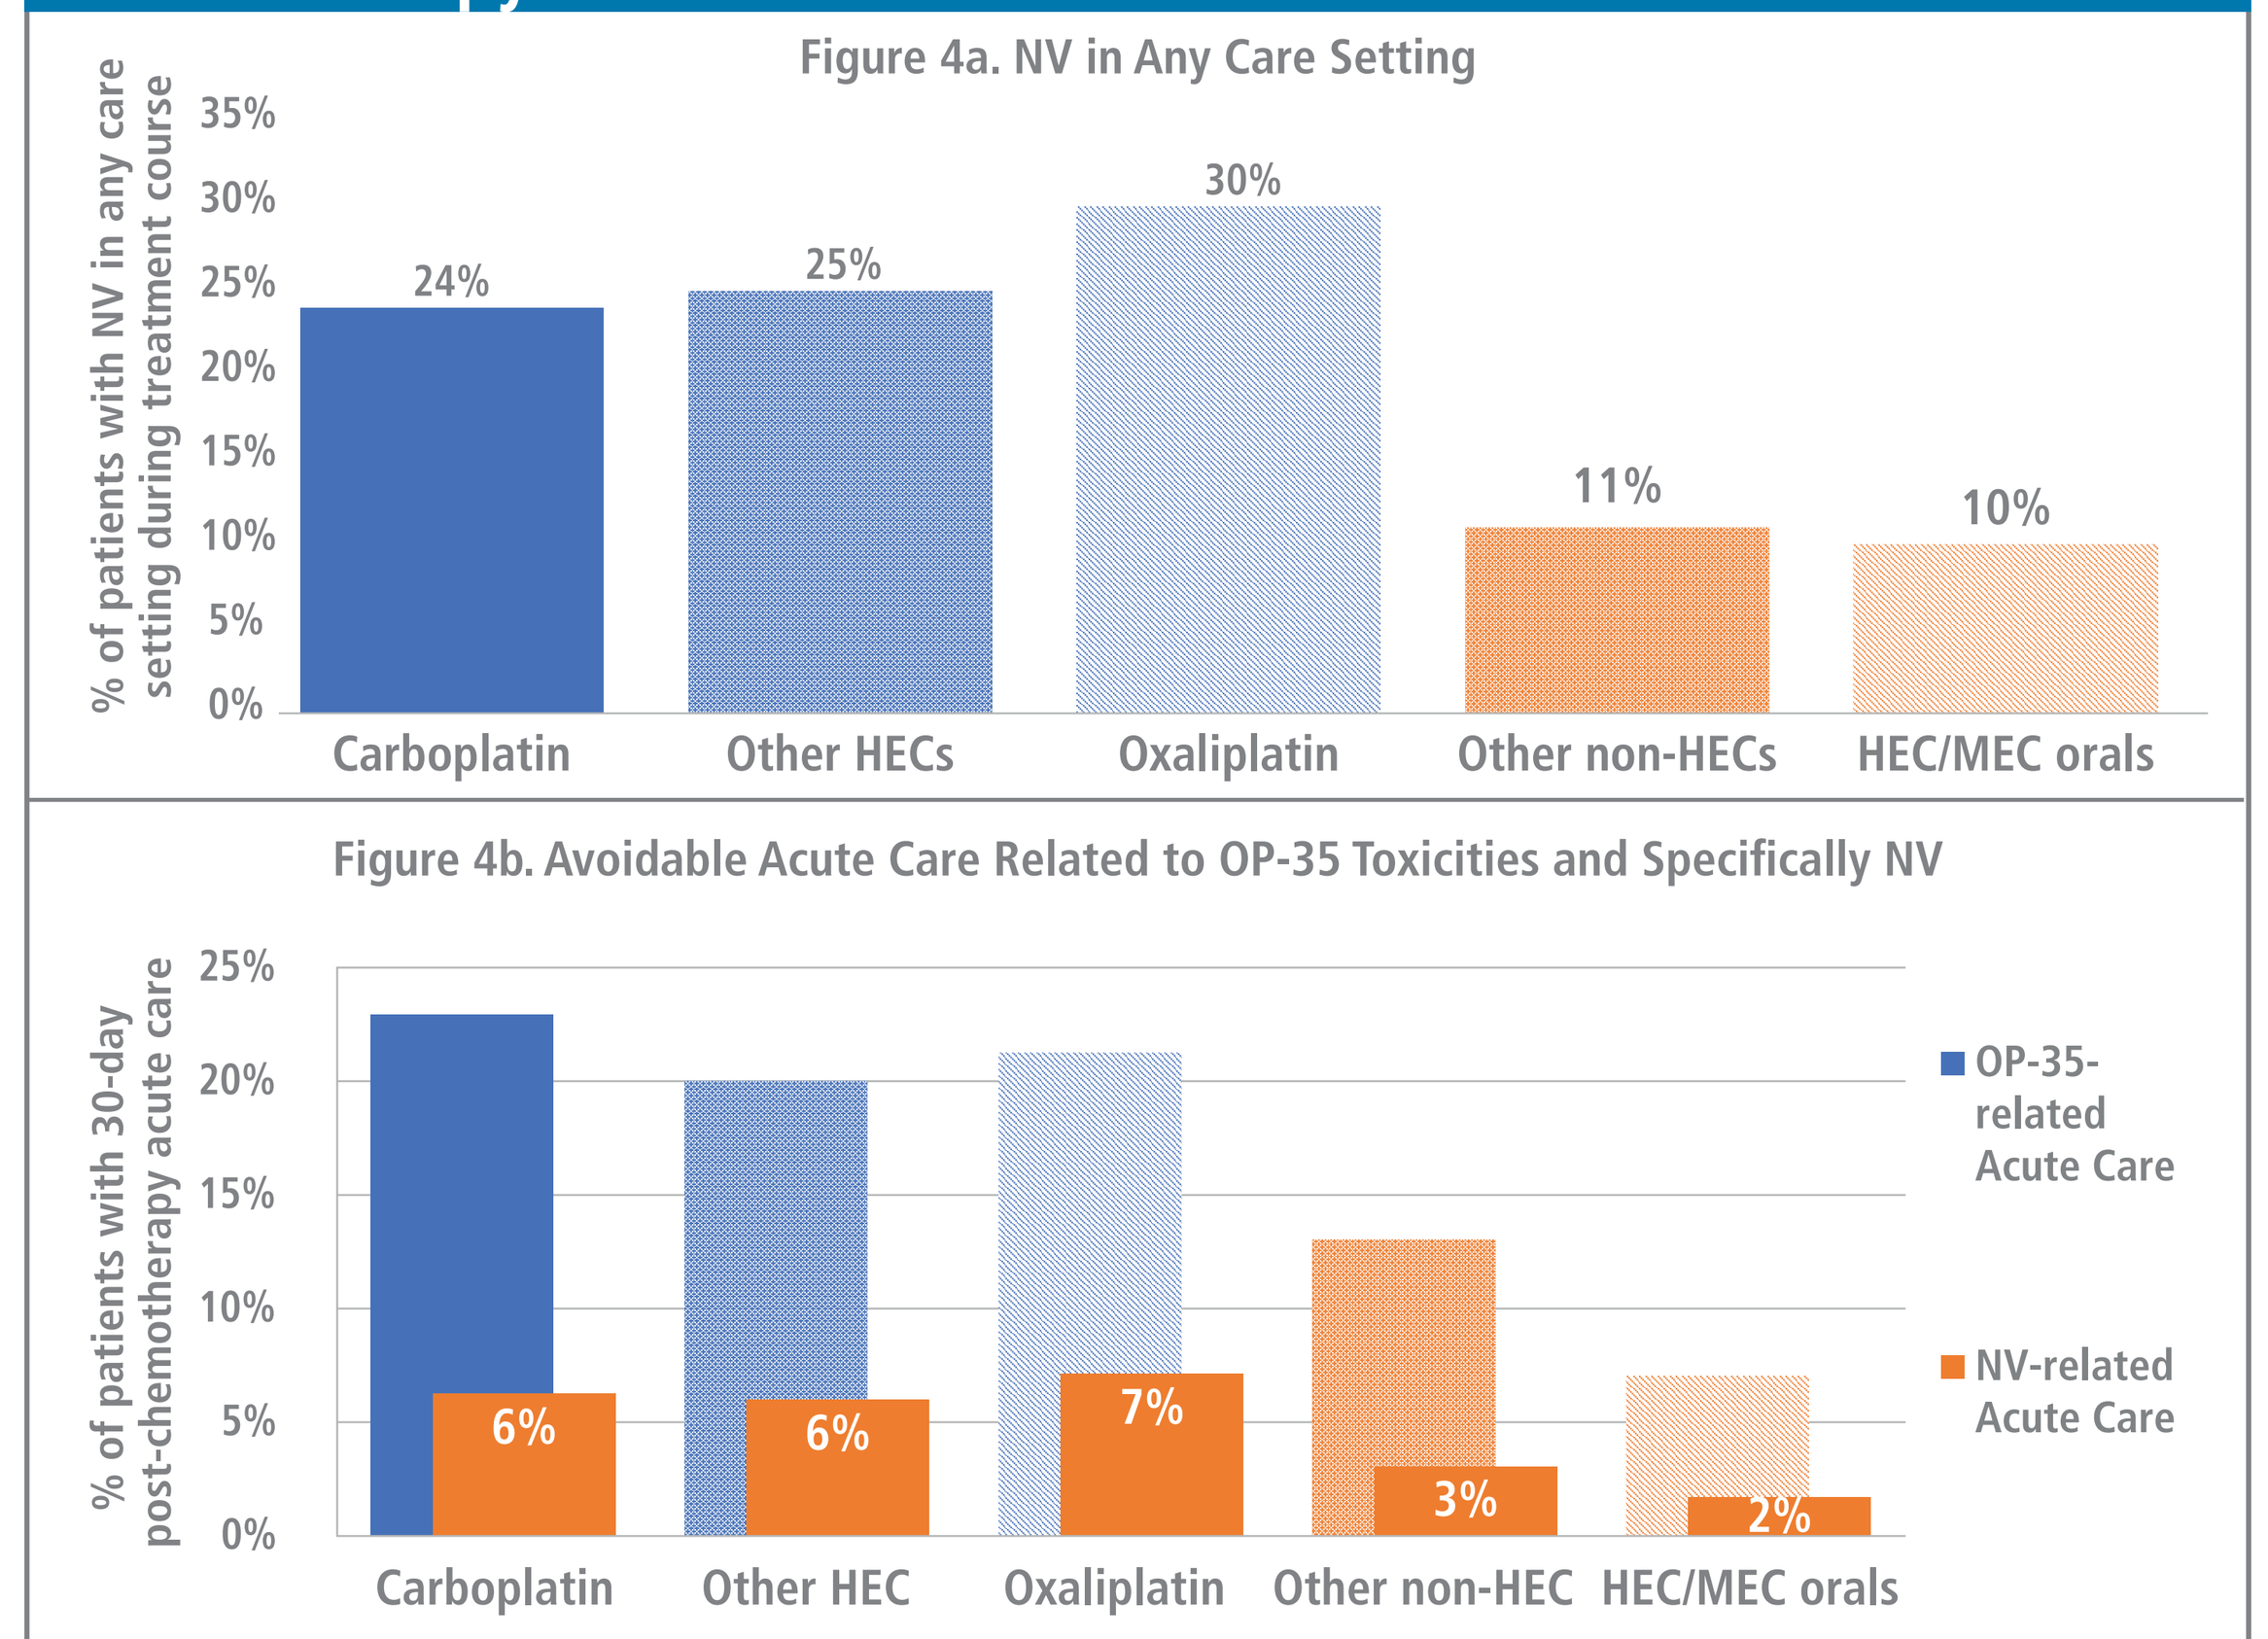
Figure 4a. NV in Any Care Setting
% of patients with NV in any care setting during treatment course
35%
30%
25%
20%
15%
10%
5%
0%
24%
25%
30%
11%
10%
Carboplatin
Other HECs
Oxaliplatin
Other non-HECs
HEC/MEC orals
Figure 4b. Avoidable Acute Care Related to OP-35 Toxicities and Specifically NV
% of patients with 30-day post-chemotherapy acute care
25%
20%
15%
10%
5%
0%
6%
6%
7%
3%
2%
Carboplatin
Other HEC
Oxaliplatin
Other non-HEC
HEC/MEC orals
OP-35-related Acute Care
NV-related Acute Care

Note: OP-35-related acute care refers to acute care events involving ≥1 of the 10 OP-35 toxicities (including NV) considered "potentially avoidable"
HEC: highly emetogenic chemotherapy; IV: intravenous; MEC: moderately emetogenic chemotherapy; NV: Nausea and vomiting

CONCLUSIONS

- Use of upfront triple antiemetic prophylaxis has increased only marginally for carboplatin AUC ≥4 since its 2017 re-classification as HEC in national guidelines, perhaps due to low awareness of the change and low awareness of the rates of post-carboplatin toxicity-related acute care.
- Patients receiving carboplatin had similar rates of NV and related 30-day acute care events as seen for other HEC, confirming that the new HEC definition fits clinical experience.
- More triple prophylaxis use is needed to reduce NV and NV-related avoidable acute care seen with carboplatin AUC ≥4.

REFERENCES

1. Berger MJ et al. *J Natl Compr Canc Netw* 2017;15:883-893
2. Hesketh PJ et al. *J of Clinical Oncol.* 2017 Oct;35(28):3240-3261.
3. Herrstedt, J, et al. *Supportive Care in Cancer* 2017;25(1): 277-288.
4. Admissions and Emergency Department Visits for Patients Receiving Outpatient Chemotherapy Measure Technical Report. Mathematica Policy Research. March 2016
5. O'Sullivan et al. *J Natl Compr Canc Netw.* 2018;16(3):294-299.
6. Roeland et al. Gaps in Compliance with Current Antiemetic Guidelines for Highly Emetogenic Chemotherapy; Poster presented at NCCN 2018.
7. Clark-Snow et al. *Support Care Cancer.* 2018;26:557-564.
8. Basch et al. *JAMA.* 2017;318(2):197-198.
9. Basch et al. *J Clin Oncol.* 2018 May;36(15):6500-6500.

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission from the authors of this poster